SPT-300, an Oral Prodrug of Allopregnanolone, Potently Reduces Salivary Cortisol Response to the Trier Social Stress Test in a Randomized, Placebo-Controlled Phase 2a Study in Healthy Participants

Michael C. Chen[1], Mark Harnett[1], Eric Elenko[2], Juliane Hellhammer[3]

[1] Seaport Therapeutics, Boston, MA, USA;
[2] PureTech Health, Boston, MA, USA;
[3] daacro, Trier, RP, Germany

SEAPORT THERAPEUTICS

Introduction

- Stress, characterized by imbalance of excitation-inhibition within corticolimbic circuits including GABAergic dysfunction, plays a key role in the pathophysiology of disorders including major depressive disorder with anxiety.[1]
- Allopregnanolone, an endogenous neurosteroid $GABA_A$ positive allosteric modulator with anti-depressant and anxiolytic activity but low oral bioavailability, is approved for the treatment of post-partum depression (PPD) as an infusion.[2]
- SPT-300* is an oral prodrug of allopregnanolone designed with the Glyph™ platform to reduce first pass metabolism in the liver and gut and allow for allopregnanolone to circulate at therapeutic levels. SPT-300 has previously shown oral bioavailability and pharmacodynamic $GABA_A$ engagement in a Phase 1 study (NCT05129865).
- To test the potential of SPT-300 to reduce physiological stress, we conducted a randomized placebo-controlled study on the effect of a single dose of SPT-300 in the Trier Social Stress Test (TSST), a clinically validated model of anxiety in healthy volunteers exposed to unpredictable, novel, anticipatory, and social stress.[3,4]

Methods

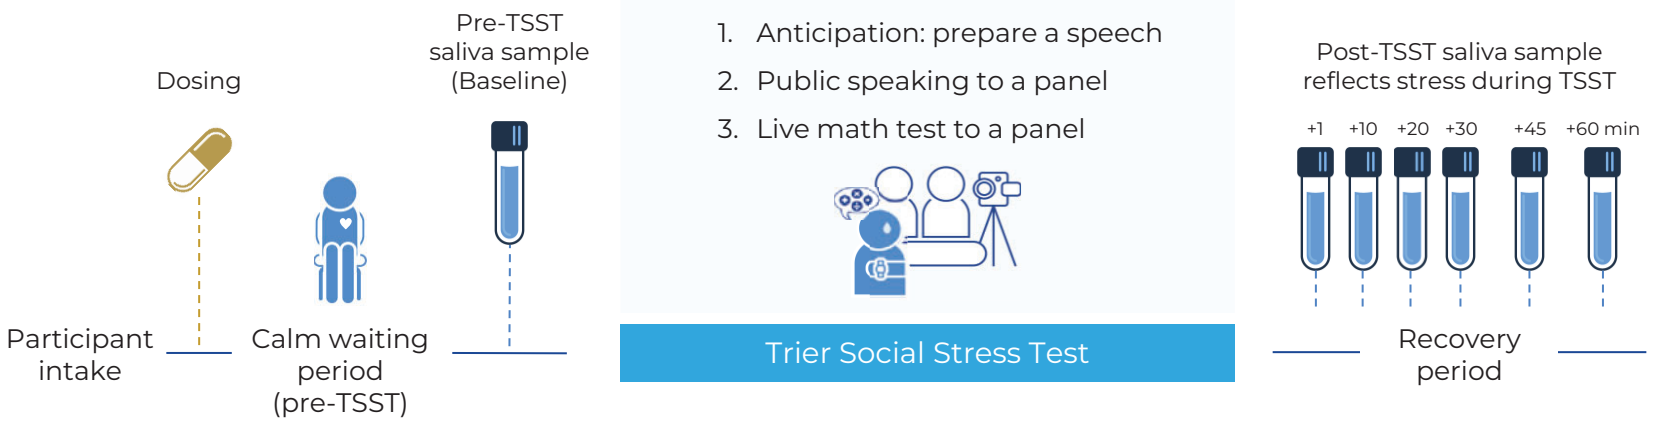
Dosing
Pre-TSST
saliva sample
(Baseline)
1. Anticipation: prepare a speech
2. Public speaking to a panel
3. Live math test to a panel
Post-TSST saliva sample
reflects stress during TSST
+1 +10 +20 +30 +45 +60 min
Participant intake
Calm waiting period (pre-TSST)
Trier Social Stress Test
Recovery period

Figure 1. TSST procedure overview

- Eighty participants were randomized to a single dose of SPT-300 (N=41) or placebo (N=39), taken prior to the TSST (NCT05129865) (Fig. 1; Table 1).
- Salivary cortisol was assessed at the time points shown above, and the primary endpoint was change from pre-treatment to the maximum cortisol value after the TSST. Cortisol values were log (base 10) transformed and analyzed with ANCOVA. Safety, tolerability, and numerical rating scales of affect were also collected.

Results

- In the placebo group, TSST substantially increased salivary cortisol, peaking at 10-20 minutes post-TSST, consistent with the known delay in cortisol response to stressor Figure 2).[4]
- SPT-300 potently blunted peak salivary cortisol response, log (base 10) transformed maximal change from baseline in salivary cortisol vs. placebo ($p = 0.0001$) (Fig. 2). The maximum non-transformed mean was also significant ($p=0.0016$) and showed SPT-300 had a ~2.2-fold decrease vs. placebo. Differences between groups ranged from 62.8% at 10-minutes post-TSST to 45.5% (60-minutes post-TSST). Across all timepoints the mean reduction in salivary cortisol was 54.4%.
- Treatment-emergent related adverse events occurring at >5% were somnolence (29% SPT-300 vs. 13% placebo) dizziness (20% SPT-300 vs. 3% placebo), and headache (7.3% SPT-300 vs. 7.7% placebo), all of which were transient and mild or moderate. No treatment-related severe or serious adverse events (AEs) were reported.
- Participants reported moderate changes in numerical ratings of affect over time, and no meaningful differences between the treatment groups or correlations between ratings and cortisol, consistent with previously reported data with TSST[3].

Variable	Statistics	Placebo (N = 39)	SPT-300 (N = 41)	Overall (N = 80)
Age (years) at Screening	n	39	41	80
	Mean (SD)	30.2 (9.6)	29.7 (10.4)	30.0 (10.0)
	Minimum	19	19	19
	Maximum	52	54	54
Sex n (%)	Female	8 (20.5%)	10 (24.4%)	18 (22.5%)
	Male	31 (79.5%)	31 (75.6%)	62 (77.5%)
Ethnicity n (%)	Hispanic or Latino	6 (15.4%)	3 (7.3%)	9 (11.3%)
	Not Hispanic or Latino	32 (82.1%)	38 (92.7%)	70 (87.5%)
	Not Reported	1 (2.6%)	0	1 (1.3%)
Race n (%)	American Indian or Alaska Native	2 (5.1%)	1 (2.4%)	3 (3.8%)
	Asian	6 (15.4%)	10 (24.4%)	16 (20.0%)
	Black or African American	0	1 (2.4%)	1 (1.3%)
	White	30 (76.9%)	29 (70.7%)	59 (73.8%)
	Other	1 (2.6%)	0	1 (1.3%)
BMI (kg/m^2) at Screening	n	39	41	80
	Mean	24.51 (2.67)	24.79 (2.37)	24.65 (2.51)
	Minimum	19.2	19.1	19.1
	Maximum	29.1	28.7	29.1

Table 1. Participant disposition

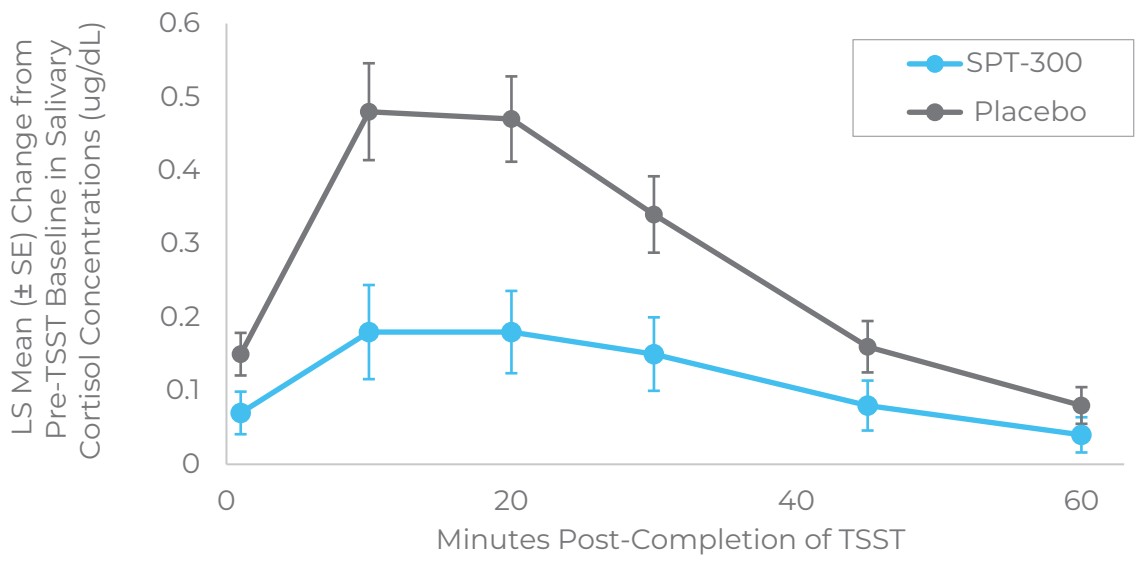
LS Mean (± SE) Change from Pre-TSST Baseline in Salivary Cortisol Concentrations (ug/dL)
0.6
0.5
0.4
0.3
0.2
0.1
0
0
20
40
60
Minutes Post-Completion of TSST
SPT-300
Placebo

Figure 2. SPT-300 blunts salivary cortisol increase to TSST

Discussion

- SPT-300 substantially reduced salivary cortisol at all post-TSST timepoints, meeting the study's primary endpoint and demonstrating that SPT-300 potently regulates hypothalamic-pituitary-adrenal axis reactivity to acute stress.
- SPT-300 was generally well-tolerated, with adverse events consistent with the pharmacological profile of allopregnanolone as well as the situational stressor of the TSST itself.
- The present study confirms SPT-300's GABA modulatory pharmacological activity, meriting further investigation in stress-related mood and anxiety disorders, including major depressive disorder with anxiety.

References:
1. Fogaça MV, Duman RS. *Frontiers in cellular neuroscience*. 2019 Mar 12;13:87.
2. Epperson CN, et al. *J. of Affective Disorders*. 2023 Jan 1;320:353-9.
3. Fries E, Hellhammer DH, Hellhammer J. *Psychoneuroendocrinology*. 2006 Nov 1;31(10):1278-88.
4. Bahr LM, et al. *Psychoneuroendocrinology*. 2021 Feb 1;124:105100.

* SPT-300 formerly known as LYT-300